ANTI-AGING EFFECT OF A LIPOHYDROXYACID AND XYLOPYRANOZIDE DERIVATIVE COMBINATION

E. Segot-Chicq, C. Laboutière, N. Caron, D. Compan-Zaouati, C. Nguyen, V. Chevalier, J. Laboureau, A. Laurent

L'Oréal Research & Innovation, Chevilly-Larue, France

INTRODUCTION

The aim of the work was to evaluate in vivo anti-ageing potential of a combination of anti ageing ingredients known to act on epidermal and dermal compartments.

MATERIALS AND METHODS

3 types of evaluations were performed on Caucasian female volunteers

1.Double blind randomized controlled clinical study on a formula containing the combination of Xylopyranozide derivate and LHA versus vehicle.

70 volunteers (45-65 y.o.) applied a formula containing the combination of 2 ingredients to one half face and vehicle on the other side, twice a day for 3 months. Self-perceived efficacy was evaluated after 3 months through a specific questionnaire.

2.Instrumental study of a skincare containing this combination together with fragmented hyaluronic acid.

42 volunteers (45-65 y.o.) applied a skincare containing the combination of ingredients (Xylopyranozide derivate + LHA + Fragmented HA), twice a day for 4 weeks.

Skin plumpness and skin density were measured using Dermotrace® and Densiscore® devices respectively at T0 and T4 weeks.

3.Clinical study of this same combination together with fragmented hyaluronic acid vs. a fractional CO_2 laser session :

2 independent groups assorted by pairs
31 volunteers (45-55 y.o.) applied a skincare containing the combination of ingredients twice daily for 8 weeks.
32 volunteers (45-55 y.o.) underwent one session of fractional CO_2 laser on the crow's feet area (ligth skin rejuvenation settings).
Imprints of the crow's feet area were taken at T0, T4 weeks and T8 weeks and analyzed by fringe projection.

1. Double blind randomized controlled clinical study on a formula containing the combination of xylopyranozide derivate and LHA vs. vehicle.

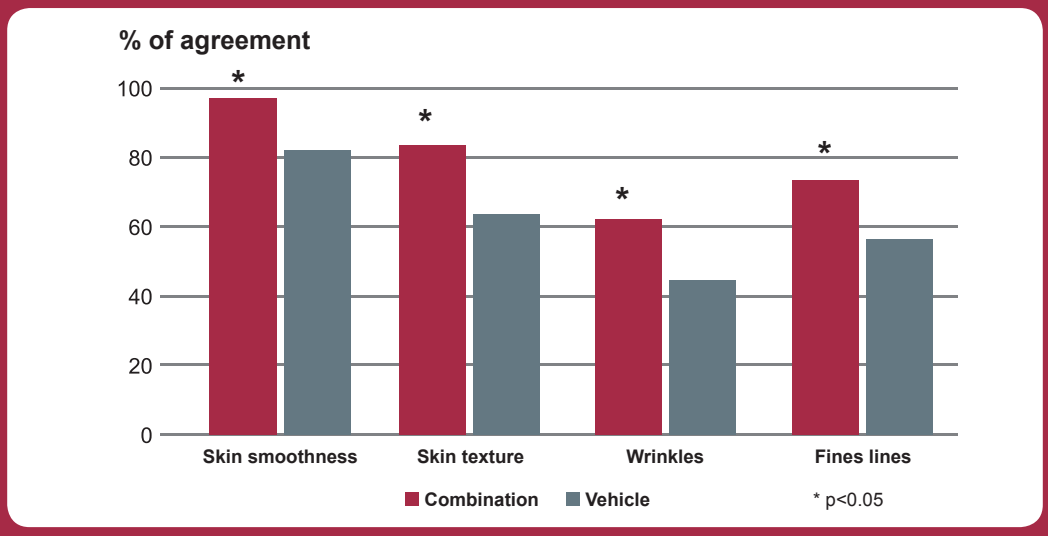
% of agreement
100
80
60
40
20
0
*
Skin smoothness
Skin texture
Wrinkles
Fines lines
Combination
Vehicle
* p<0.05

Perceived efficacy after 3 months usage on skin smoothness and skin texture, on wrinkles and fine lines was significantly better with the combination than vehicle.

2. Instrumental study of a skincare containing this combination and fragmented hyaluronic acid.

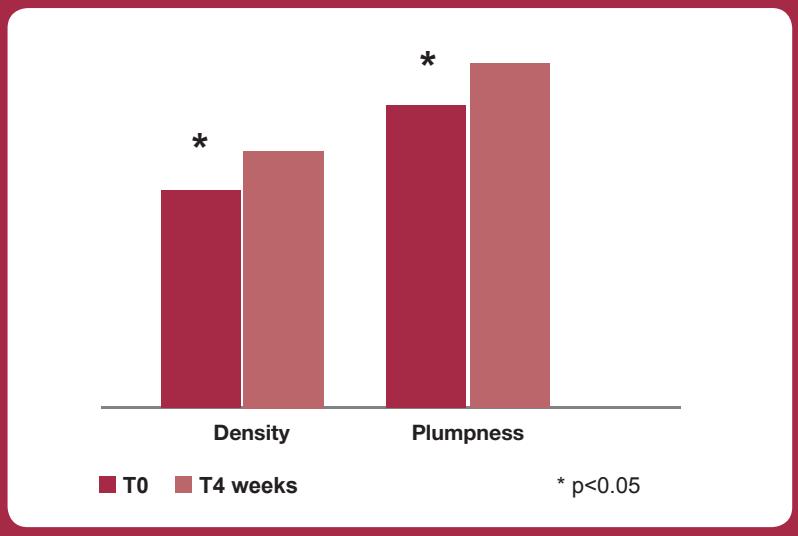
*
Density
Plumpness
T0
T4 weeks
* p<0.05

Instrumental evaluation after 4 weeks showed significant improvement of skin plumpness (by 12%) and of skin density (by 11%) vs base line.

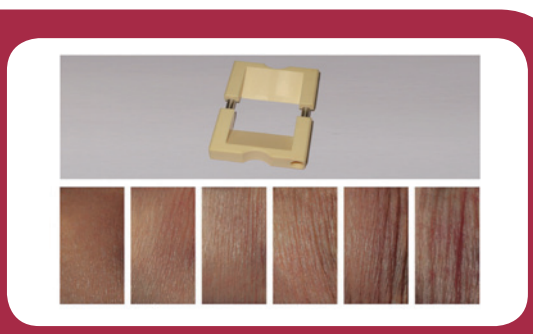

Densicore

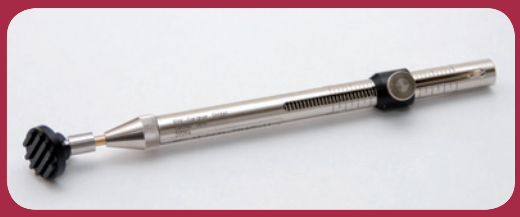

Dermotrace

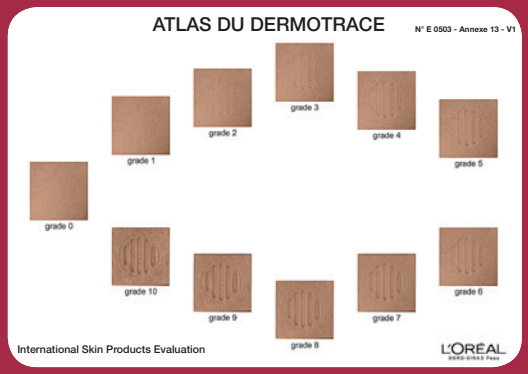
ATLAS DU DERMOTRACE
International Skin Products Evaluation

3. Clinical study of a skincare containing the combination of xylopyranoside and LHA together with fragmented hyaluronic acid vs. a fractional CO_2 laser session
Evaluation by fringe projection on the crow's feet area (imprints)

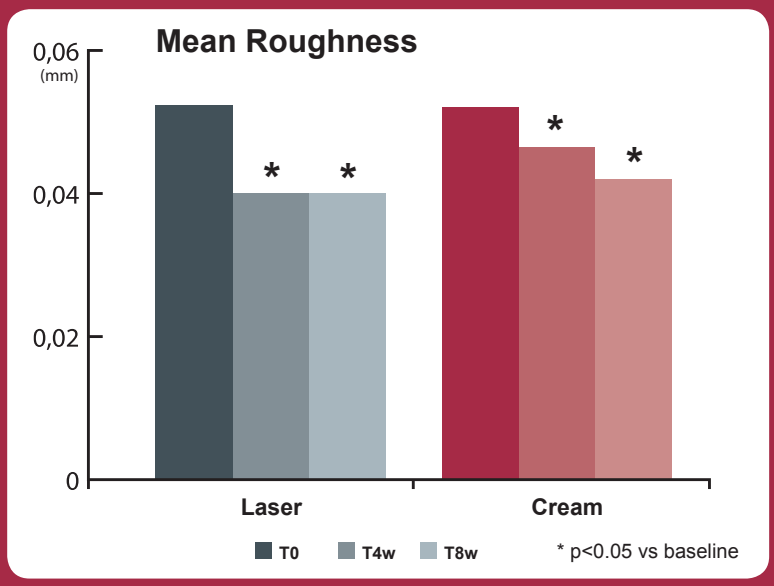
Mean Roughness
0,06
(mm)
0,04
0,02
0
*
Laser
Cream
T0
T4w
T8w
* p<0.05 vs baseline

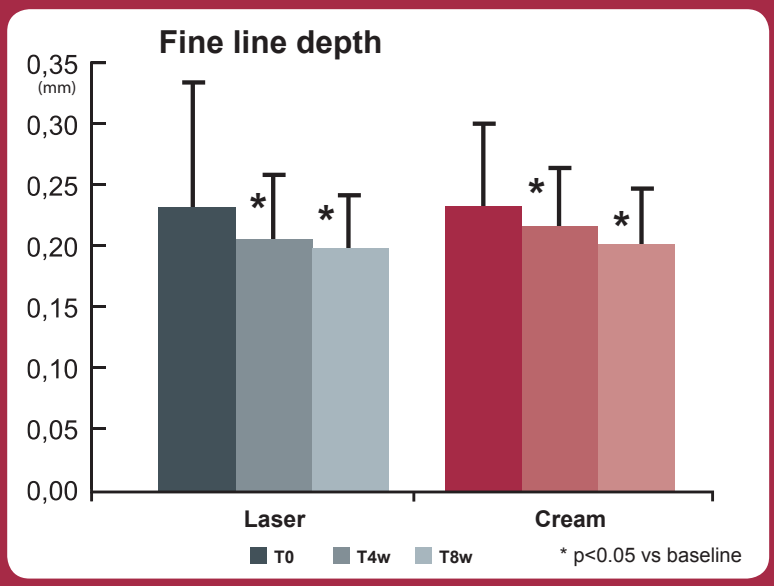
Fine line depth
0,35
(mm)
0,30
0,25
0,20
0,15
0,10
0,05
0,00
*
Laser
Cream
T0
T4w
T8w
* p<0.05 vs baseline

VISIBLE REDUCTION OF THE CROW'S FEET

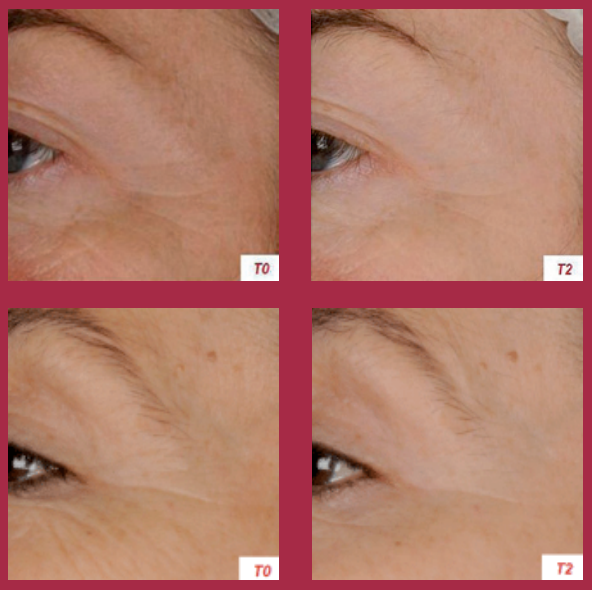
T0
T2
T0
T2

One subject at T0 after one session of fractionnal CO_2 laser.

One subject at T0 and after 8 weeks of twice daily use of the skincare.

SIGNIFICANT IMPROVEMENT OF SKIN MICROSURFACE WAS OBSERVED AFTER 8 WEEKS for both groups, with no statistical difference between the cream and the laser session (fractional CO_2 laser, settings for light skin rejuvenation)

CONCLUSION

The Xylopyranoside derivative and Lipo-hydroxacid combination showed enhanced perception of efficacy on skin smoothness, fine lines and wrinkles vs. vehicle.
Instrumental measurements gave evidence that skin density and skin plumpness were clearly improved vs. baseline by a skincare containing this combination and fragmented hyaluronic acid.
Application of the skincare containing this combination of actives ingredients and fragmented hyaluronic acid twice a day for 8 weeks showed smoothing efficacy on the crow's feet area not statistically different to one single CO_2 fractioned laser session (light skin rejuvenation settings).

L'ORÉAL PARIS

21st EADV Congress, Prague, Czech Republic, September 2012

L'ORÉAL Research & Innovation